Discovery of Helicon Peptides for the Selective Degradation of the Agonist-Bound Conformation of Androgen Receptor (AR^{ON}) in Prostate Cancer

Diwakar R. Pattabiraman*, Brandon Nicolay*, Pieter Beerepoot, Kelsey Barrasso, Sascha DeVine, Mandana Abbassi, Terri Jones, Dakota Hawkins, Archana Iyer, Lee Belding, Shelagh Fluharty, Amelia K. Luciano, Paula Ortet, Anthony Quartararo, Yelena Arnautova, David Terry, Brandon Hriniak, Mousa Jafari, Donovan Chin, Anandan Palani, Markus Haeberlein, Jonathan Hurov

Parabilis Medicines Inc., 30 Acorn Park Drive, Cambridge MA

Background

The androgen receptor (AR) is a critical driver of prostate cancer, regulating genes that control tumor growth, survival, and progression. Androgen deprivation therapy (ADT) is a cornerstone of treatment, but many patients eventually develop castration-resistant prostate cancer (CRPC), in which AR signaling remains active through mutations, amplification, or splice variants. Thus, AR remains a prime therapeutic target, and drug discovery has focused on next-generation AR antagonists, degraders, and other approaches to overcome resistance. Advances in targeting AR biology continue to shape the landscape of prostate cancer therapy and drive innovation in precision oncology.

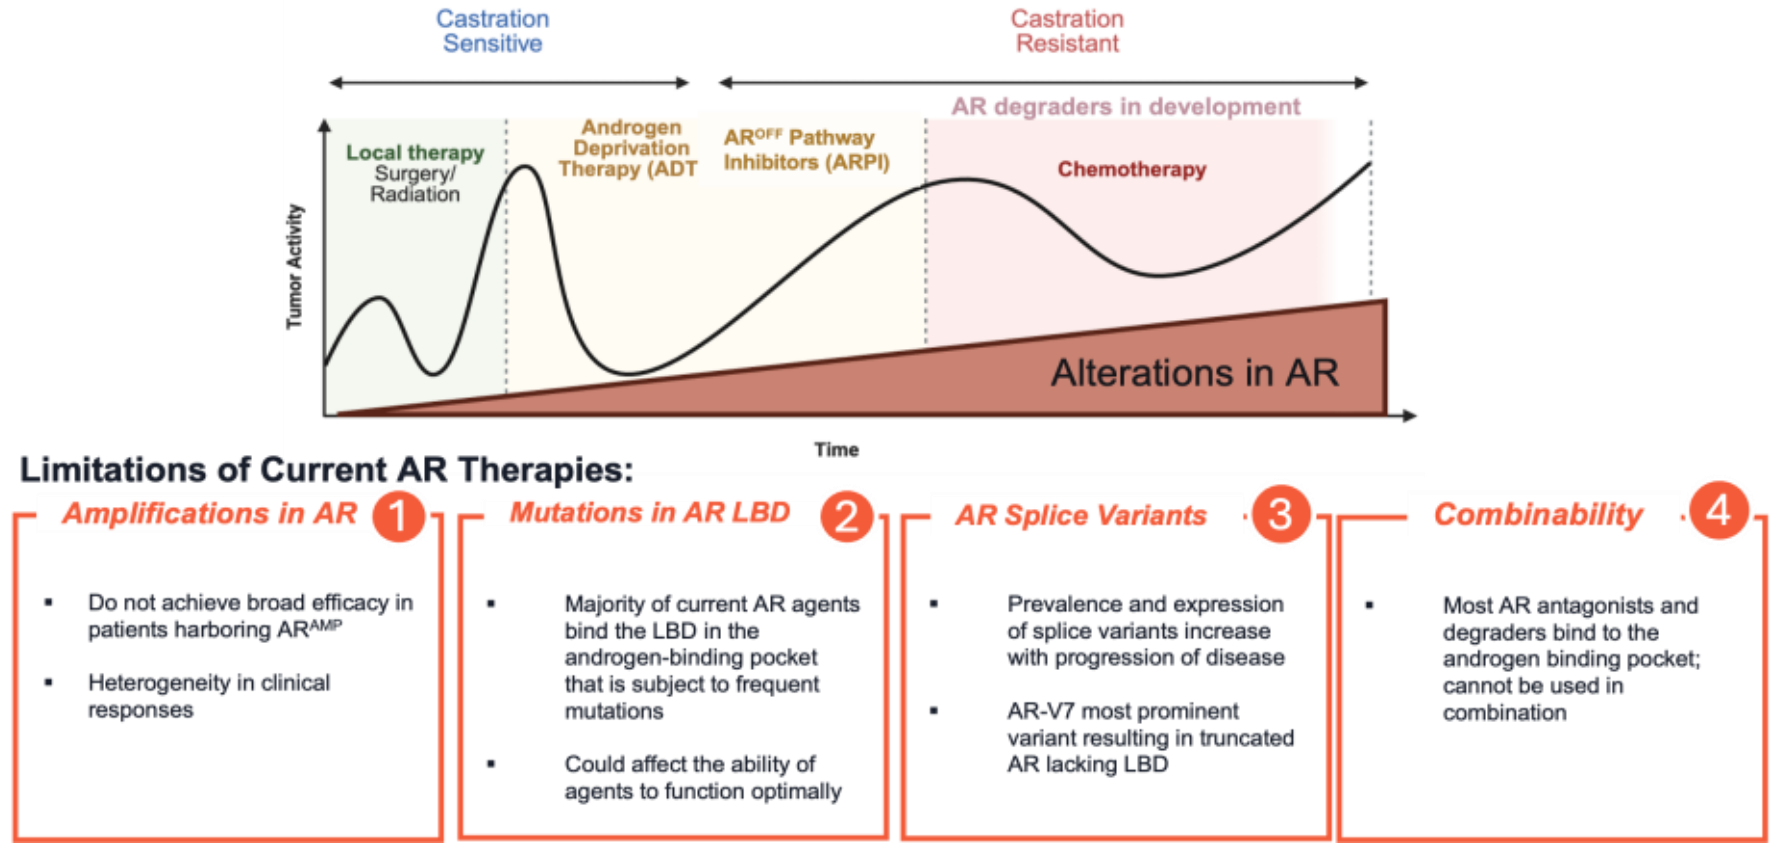
Castration Sensitive
Castration Resistant
AR degraders in development
Local therapy Surgery/ Radiation
Androgen Deprivation Therapy (ADT
AR^OFF Pathway Inhibitors (ARPI)
Chemotherapy
Tumor Activity
Alterations in AR
Time
Limitations of Current AR Therapies:
Amplifications in AR 1
Do not achieve broad efficacy in patients harboring AR^AMP
Heterogeneity in clinical responses
Mutations in AR LBD 2
Majority of current AR agents bind the LBD in the androgen-binding pocket that is subject to frequent mutations
Could affect the ability of agents to function optimally
AR Splice Variants 3
Prevalence and expression of splice variants increase with progression of disease
AR-V7 most prominent variant resulting in truncated AR lacking LBD
Combinability 4
Most AR antagonists and degraders bind to the androgen binding pocket; cannot be used in combination

Currently approved agents and the majority of those in development bind to the inactive conformation of AR that is not androgen bound (AR^{OFF}), preventing activation of the protein, rather than targeting it in the active, agonist-bound conformation that is responsible for driving transcription and tumorigenesis. Tumors that retain AR pathway dependence while treated with these inhibitors respond with adaptive activation of AR signaling, which in turn is associated with relapse and disease progression.

Our Unique Approach to Targeting AR

Our Helicon-based bifunctional degraders bind to the AR LBD in its agonist-bound conformation, at a site distinct from the ligand binding pocket targeted by AR^{OFF} agents. Binding to a unique LBD site outside of ligand binding pocket enables us to selectively degrade the AR^{ON} agonist-bound pool.

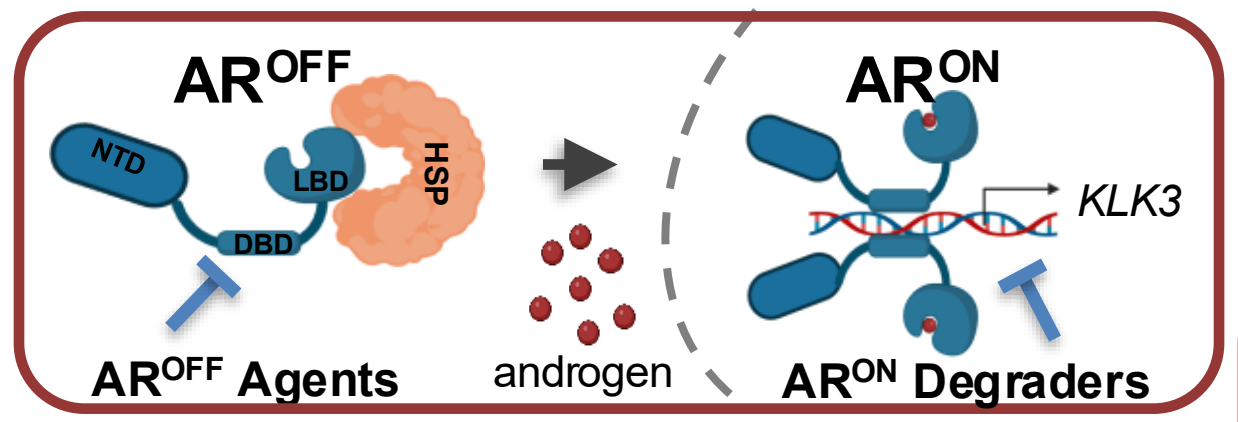
AR^OFF
NTD
LBD
DBD
HSP
AR^ON
KLK3
AR^OFF Agents
androgen
AR^ON Degraders

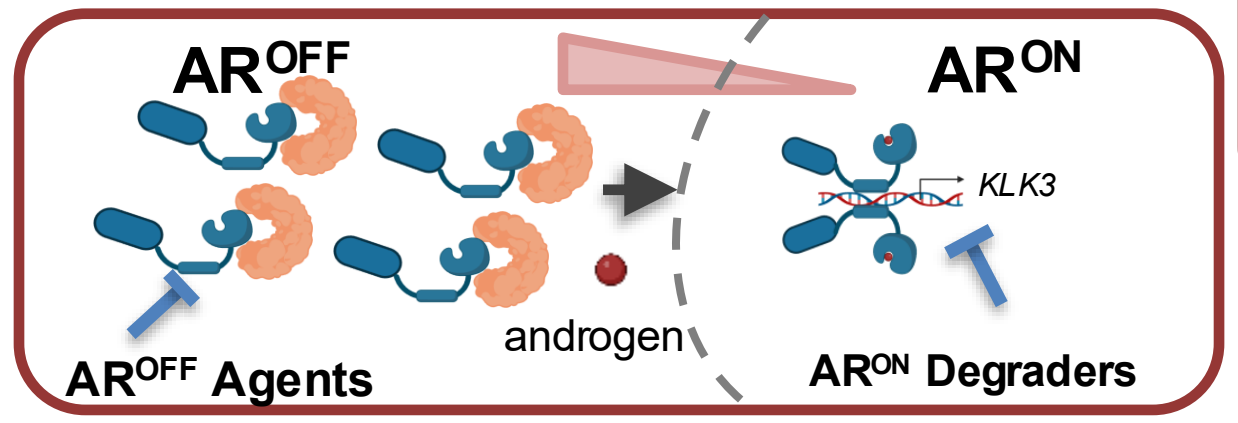
AR^OFF
AR^ON
KLK3
androgen
AR^OFF Agents
AR^ON Degraders

Key advantages of AR^{ON} Helicon-degraders:
- Known LBD point mutations do not alter Helicon binding
- Amplified AR does not act as a decoy
- Ability to combine with current SoC AR^{OFF} agents

Computational *De Novo* Discovery and Optimization

Using a combination of *de novo* computational and structure-based molecular design, we discovered compounds that bind to the AR LBD in its agonist-bound conformation, at a site distinct from the ligand binding pocket targeted by AR^{OFF} agents.

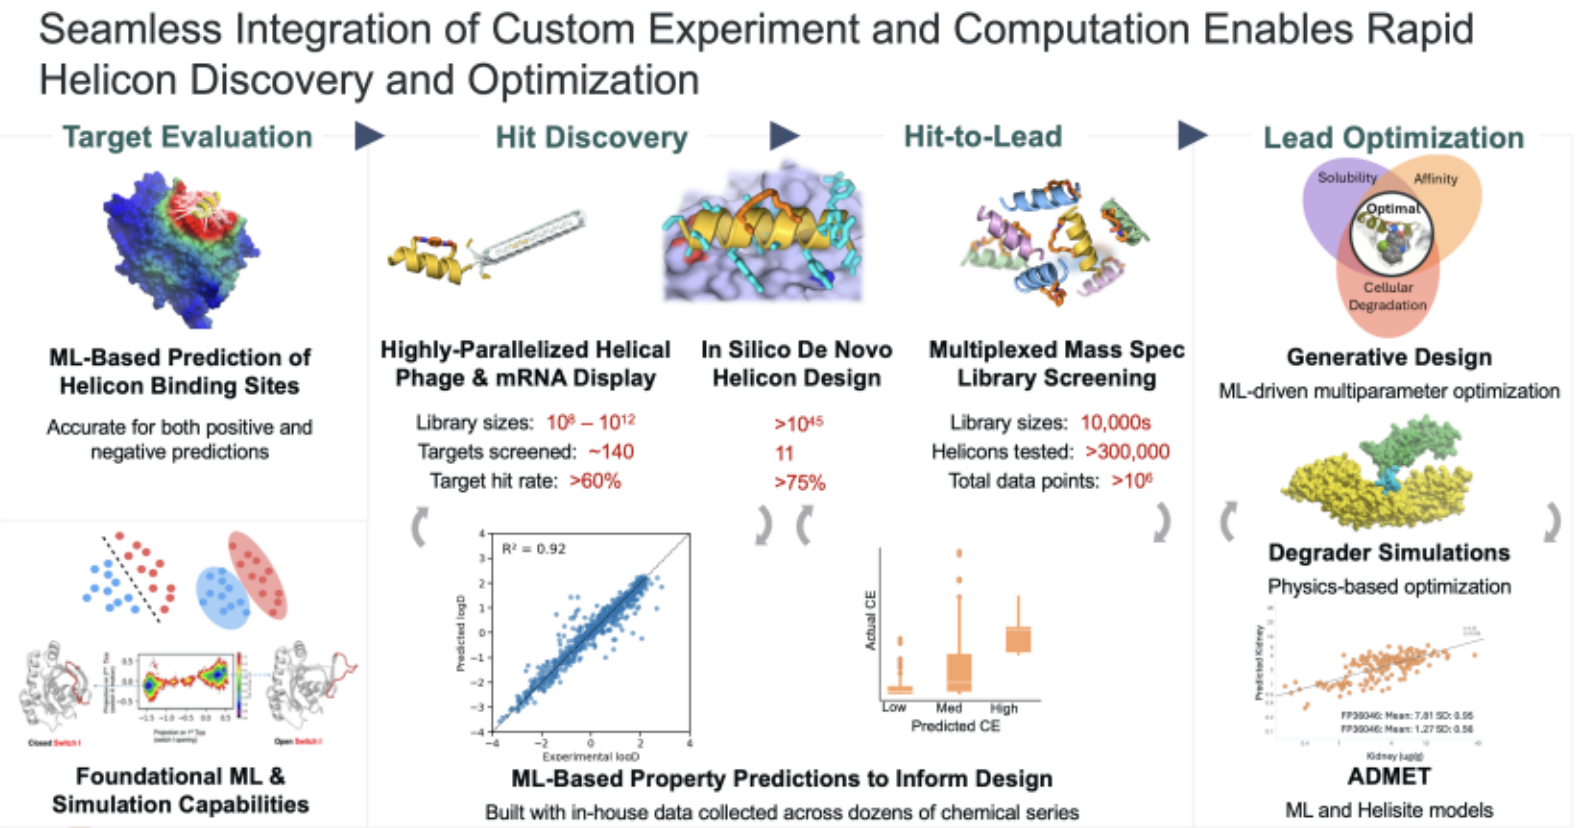
Seamless Integration of Custom Experiment and Computation Enables Rapid Helicon Discovery and Optimization
Target Evaluation
Hit Discovery
Hit-to-Lead
Lead Optimization
ML-Based Prediction of Helicon Binding Sites
Accurate for both positive and negative predictions
Highly-Parallelized Helical Phage & mRNA Display
Library sizes: 10^8 – 10^12
Targets screened: ~140
Target hit rate: >60%
In Silico De Novo Helicon Design
>10^45
11
>75%
Multiplexed Mass Spec Library Screening
Library sizes: 10,000s
Helicons tested: >300,000
Total data points: >10^6
Solubility
Affinity
Optimal
Cellular Degradation
Generative Design
ML-driven multiparameter optimization
Degrader Simulations
Physics-based optimization
Foundational ML & Simulation Capabilities
R² = 0.92
Actual CE
Low
Med
High
Predicted CE
ML-Based Property Predictions to Inform Design
Built with in-house data collected across dozens of chemical series
ADMET
ML and Helisite models

- Customized high-throughput chemistry on 1000's of non-natural amino acids and 100's of staples
- Proprietary high-quality data from *in vitro* and *in vivo* biological data
- Custom AI/ML models and physics-based modeling connects biology to Helicon binding sites on targets of interest and to navigate vast chemical space

AR^{ON} Helicons Bind to Agonist-bound LBD

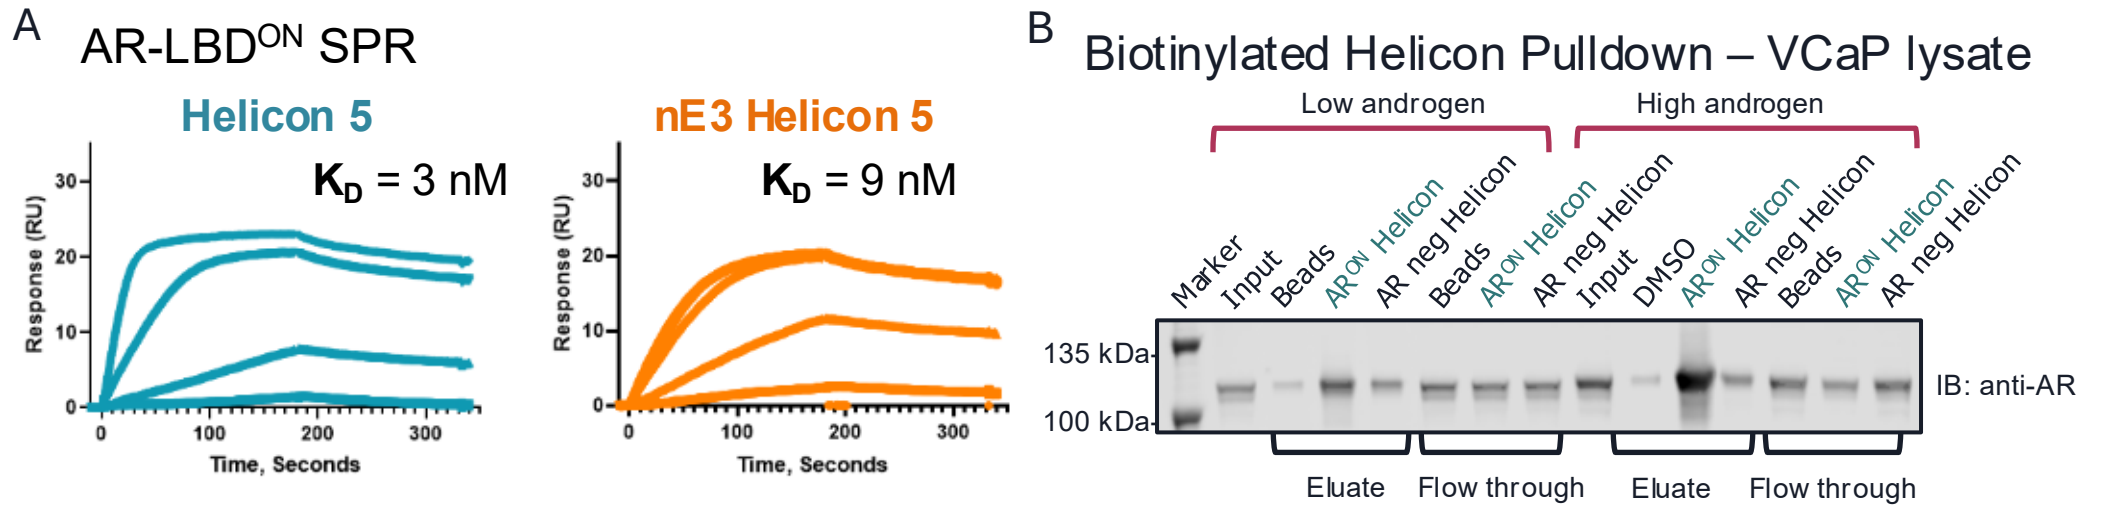
A
AR-LBD^ON SPR
Helicon 5
K_D = 3 nM
nE3 Helicon 5
K_D = 9 nM
Response (RU)
Time, Seconds
B
Biotinylated Helicon Pulldown – VCaP lysate
Low androgen
High androgen
Marker
Input
Beads
AR^ON Helicon
AR neg Helicon
DMSO
135 kDa
100 kDa
IB: anti-AR
Eluate
Flow through

Figure 1. AR^{ON} helicons bind to agonist bound AR LBD. (A) AR^{ON} helicons bind recombinant, agonist-loaded LBD with high affinity as measured by SPR (B) AR pulldown by biotinylated helicons from VCaP lysate is strongly increased by treating cells in culture with high androgen concentration prior to generating lysate.

Degradation of AR^{ON} Decreases AR Signaling and Cell Proliferation

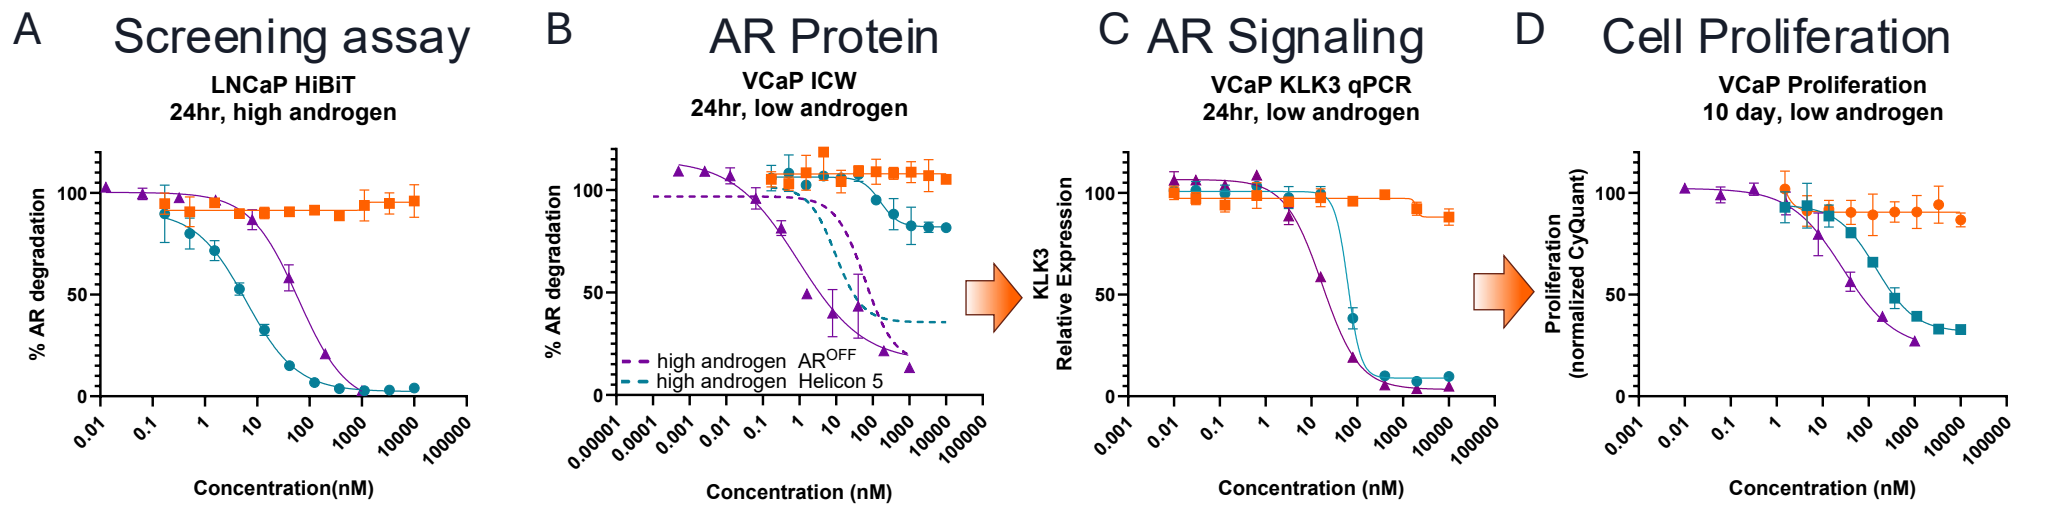
A Screening assay
LNCaP HiBiT
24hr, high androgen
% AR degradation
Concentration(nM)
B AR Protein
VCaP ICW
24hr, low androgen
high androgen AR^OFF
high androgen Helicon 5
C AR Signaling
VCaP KLK3 qPCR
24hr, low androgen
KLK3 Relative Expression
D Cell Proliferation
VCaP Proliferation
10 day, low androgen
Proliferation (normalized CyQuant)
Concentration (nM)

A	DC50 (nM)	Dmax
Helicon 5	6	96%
nE3 Helicon 5	>10000	11%
AR^{OFF}	61	97%

B	DC50 (nM)	Dmax
Helicon 5	164	18%
nE3 Helicon 5	>10000	0%
AR^{OFF}	1	86%

C	IC50 (nM)	Emax
Helicon 5	63	91%
nE3 Helicon 5	>10000	12%
AR^{OFF}	17	95%

D	GI50 (nM)	Emax
Helicon 5	145	67%
nE3 Helicon 5	>10000	13%
AR^{OFF}	28	72%

Figure 2. AR^{ON} helicons are nanomolar potency AR degraders that reduce AR dependent transcription and cell proliferation in VCaP cells. (A) AR degradation in LNCaP cell lines expressing AR-HiBiT (10nM R1881) and (B) VCaP In Cell Western (no added androgen) (C). KLK3 transcript levels measured by qPCR (D) Cell proliferation after 10 days as measured by DNA-content using CyQUANT (no added androgen).

AR^{ON} Helicons Preferentially Reduce Nuclear AR

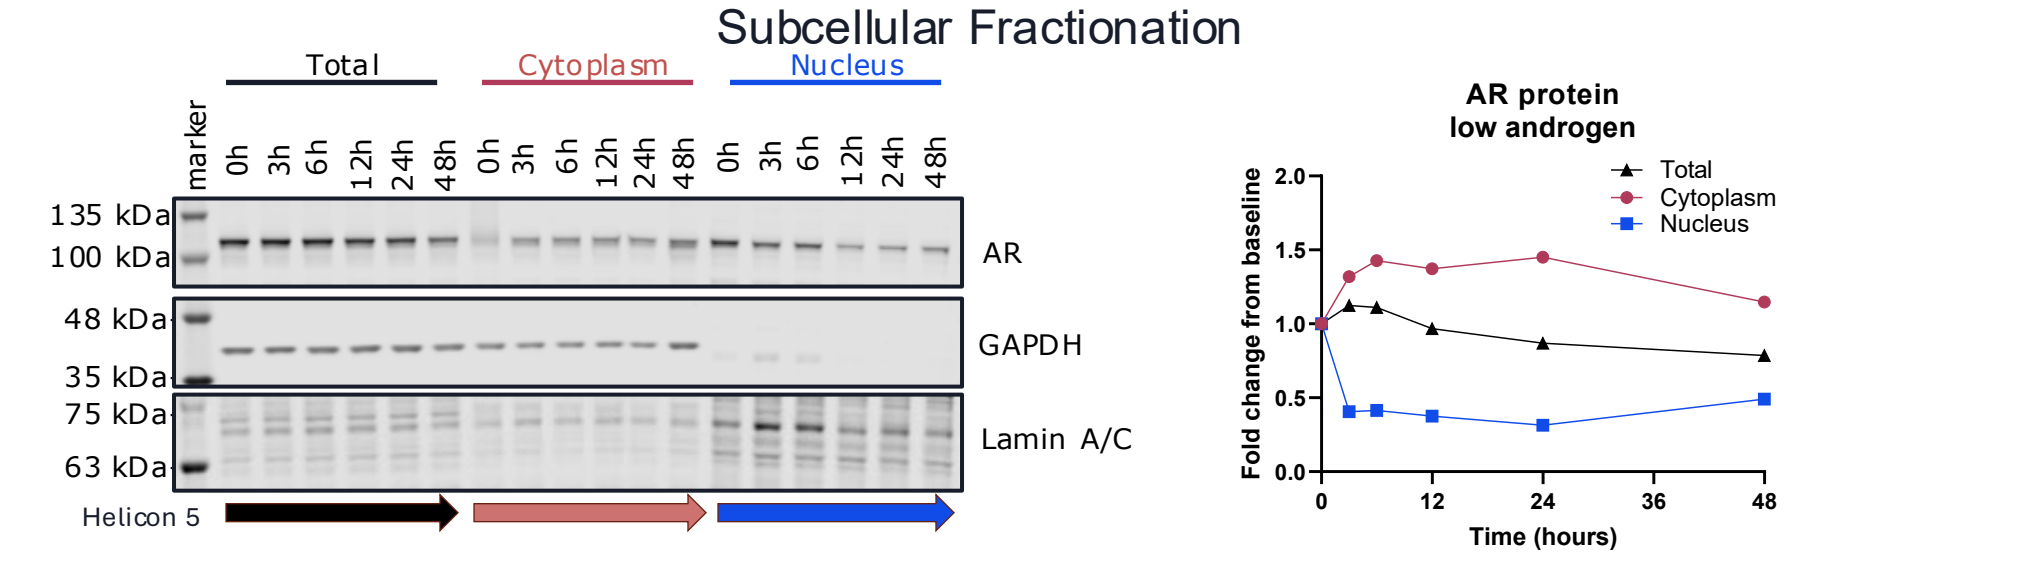
Subcellular Fractionation
Total
Cytoplasm
Nucleus
marker
0h 3h 6h 12h 24h 48h
135 kDa
100 kDa
AR
48 kDa
35 kDa
GAPDH
75 kDa
63 kDa
Lamin A/C
Helicon 5
AR protein
low androgen
Fold change from baseline
Time (hours)

Figure 3. Subcellular fractionation of VCaP cells treated with AR^{ON} degrader demonstrates preferential degradation of nuclear pool of AR

AR^{ON} Degraders results in selective, E3/UPS-dependent AR degradation

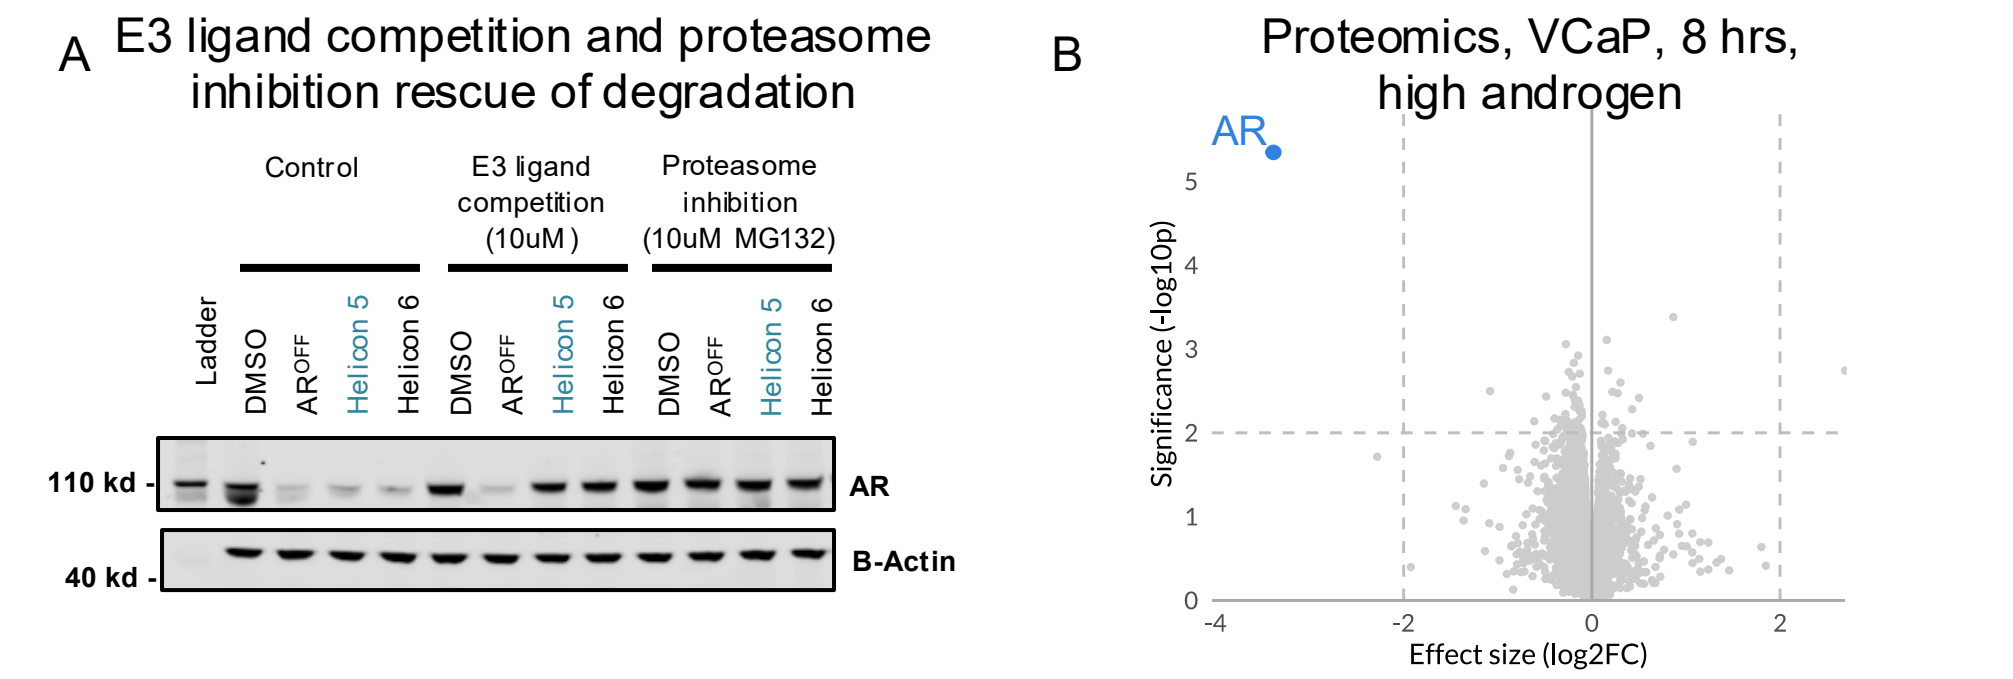
A
E3 ligand competition and proteasome inhibition rescue of degradation
Control
E3 ligand competition (10uM)
Proteasome inhibition (10uM MG132)
Ladder
DMSO
AR^OFF
Helicon 5
Helicon 6
110 kd
AR
40 kd
B-Actin
B
Proteomics, VCaP, 8 hrs, high androgen
AR
Significance (-log10p)
Effect size (log2FC)

Figure 4. AR^{ON} helicons selectively degrade AR in E3 ligase and UPS-dependent manner in LNCaP cells (A) Competition with 10x excess E3 ligand blocks helicon or proteasome inhibition with MG132 block helicon degrader activity. (B) Proteomics profiling comparing helicon 5 with vehicle control

AR^{ON} degraders demonstrate combination potential with ARPI SoC

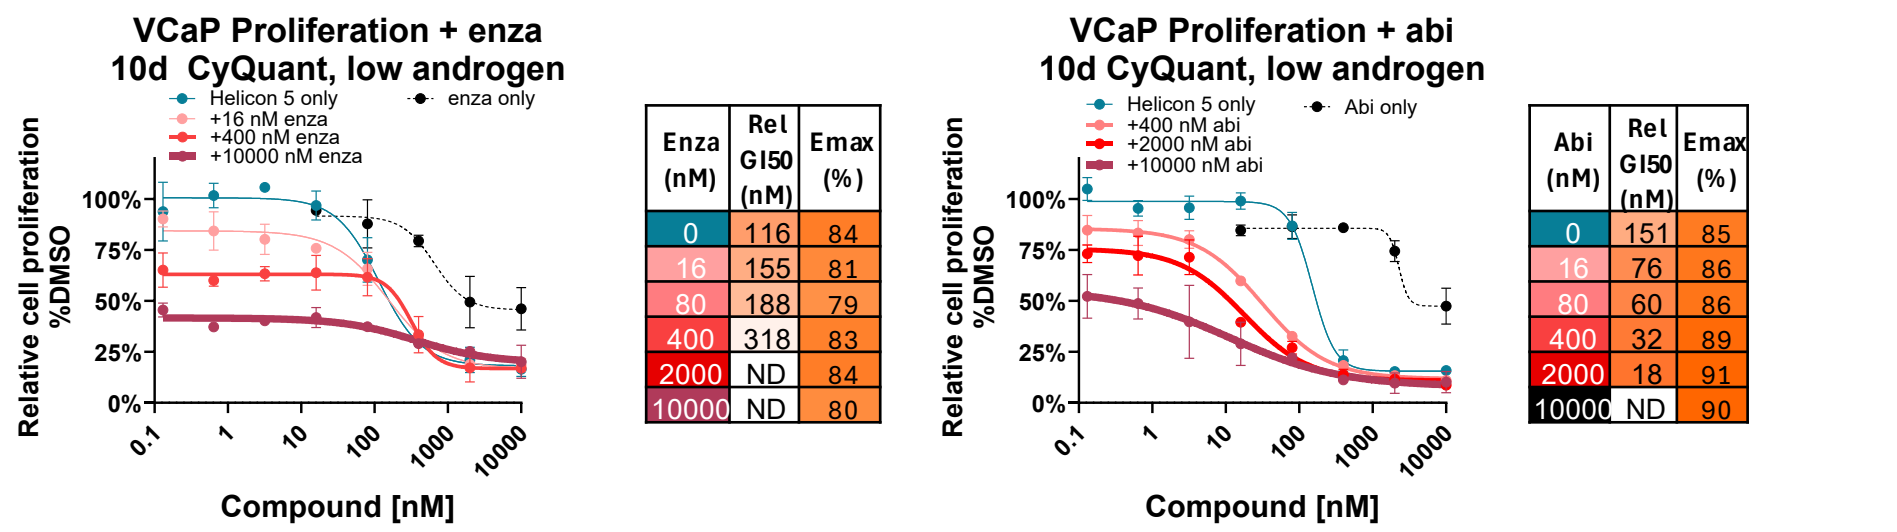
VCaP Proliferation + enza
10d CyQuant, low androgen
Helicon 5 only
+16 nM enza
+400 nM enza
+10000 nM enza
enza only
Relative cell proliferation %DMSO
Compound [nM]
VCaP Proliferation + abi
10d CyQuant, low androgen
Helicon 5 only
+400 nM abi
+2000 nM abi
+10000 nM abi
Abi only

Enza (nM)	Rel GI50 (nM)	Emax (%)
0	116	84
16	155	81
80	188	79
400	318	83
2000	ND	84
10000	ND	80

Abi (nM)	Rel GI50 (nM)	Emax (%)
0	151	85
16	76	86
80	60	86
400	32	89
2000	18	91
10000	ND	90

Figure 5. AR^{ON} helicons were combined with either enzalutamide or abiraterone in 10-day CyQuant proliferation assay. Enzalutamide had additive effects to helicon across most of the concentration range. Abiraterone combination had synergistic effects, and shifted the helicon dose response left

Conclusions

- We have developed Helicon-based bifunctional degraders of the AR^{ON} pool of the androgen receptor with potent binding affinity and degradation
- Our Helicon-based degraders are E3-dependent and selective for AR
- AR^{ON} degradation is sufficient to inhibit AR signaling and cell proliferation, despite constituting only a portion of total AR under low androgen conditions
- *In vitro* evidence suggests AR^{ON} degraders can be combined with SoC ARPIs such as enzalutamide and abiraterone without observing antagonistic effects

Acknowledgments

We acknowledge the support of Pharmaron Inc., Viva, Helix, Wuxi and Molsoft for their contributions to our AR program

Presented at the AACR-NCI-EORTC International Conference on Molecular Targets and Cancer Therapeutics 2025